Mortality among veterans with a diagnosis of pyruvate kinase deficiency: A real-world study using US Veterans Health Administration data

e-Poster # EP710

Erin Zagadailov, PharmD, MS[1] Audra N Boscoe, PhD,[1] Viviana Garcia-Horton, PhD,[2] Sherry Shi, MSc,[3] Shuqian Liu, MD,[4] Lizheng Shi, PhD,[4] Dendy Macaulay, PhD[2]

[1]Agios Pharmaceuticals, Inc., Cambridge, MA, United States; [2]Analysis Group, Inc., New York, NY, United States; [3]Analysis Group, Inc., Montreal, QC, Canada; [4]School of Public Health & Tropical Medicine, Tulane University, New Orleans, LA, United States

BACKGROUND

- Pyruvate kinase (PK) deficiency is a rare, underrecognized inherited disorder caused by autosomal recessive mutations in the *PKLR* gene
 - The *PKLR* gene encodes red blood cell (RBC) PK (PKR), which is critical for maintaining RBC energy levels and morphology[1,2]
 - A glycolytic defect causes reduced adenosine triphosphate levels and leads to hemolytic anemia[1,2]
 - Defects in PKR cause chronic hemolysis, which leads to other long-term complications[3,4]
 - These include gallstones, pulmonary hypertension, extramedullary hematopoiesis, osteoporosis, and iron overload and its sequelae[3–5]
 - PK deficiency can also cause quality of life problems, including challenges with work and school activities, social life, and emotional health[6]
- Current management strategies for PK deficiency, including RBC transfusions and splenectomy, are associated with both short- and long-term risks[3,7]
- Population-based studies of PK deficiency using claims or electronic health record databases are limited
 - Identifying PK deficiency in real-world data is challenging due to a lack of diagnosis codes and treatments that are specific to the disease
- Data on mortality in this patient population are lacking and limited to a few individual case reports[8–16]
- The US Veterans Health Administration (US VHA) database was selected for this research because of its long length of follow-up and availability of death data

OBJECTIVES

- Identify patients with a PK deficiency diagnosis as documented by physicians
- Compare their rates of mortality to an age- and gender-matched cohort of individuals without PK deficiency

METHODS

PK deficiency cohort

- Patients with ≥ 1 diagnosis code related to PK deficiency between January 1995 and July 2019 were selected from the US VHA database
 - Anemia due to disorders of glycolytic enzymes (ICD-10-CM: D55.2), other hemolytic anemias due to enzyme deficiency (ICD-9-CM 282.3), or unspecified hereditary hemolytic anemia (ICD-9-CM 282.9, ICD-10-CM D58.9)
- To be considered for inclusion in this research, physicians' notes were required to contain the words "pyruvate", "kinase", and "deficiency"
- A manual review of these physicians' notes was performed to identify patients with a physician-documented diagnosis of PK deficiency
- The index date for the PK deficiency cohort was defined as the date of the first medical record with a diagnosis code related to PK deficiency

Non-PK deficiency cohort

- Each patient in the PK deficiency cohort was matched 1:5 by age at index, sex, and index year (± 1 year) to patients from the general US VHA population with no diagnosis codes related to PK deficiency
- The index date for the non-PK deficiency cohort was defined as a random visit date during their match's index year

Patient characteristics

- Demographic and clinical characteristics were compared between the PK deficiency cohort and their non-PK deficiency cohort matches

Survival analysis

- Survival time from the index date between the PK deficiency cohort and their non-PK deficiency cohort matches was summarized using Kaplan–Meier survival estimates and compared using a univariate Cox proportional hazards model with robust standard error estimation

RESULTS

Patient characteristics

- A total of 18 patients met inclusion criteria for the PK deficiency cohort and were matched to 90 individuals in the non-PK deficiency cohort
- Baseline characteristics for both cohorts are shown in **Table 1**
- For both cohorts, the mean age at index was 56.8 years
- There were no significant differences between age, sex, race, or mean Charlson Comorbidity Index
- Imbalances remained between the two cohorts with regard to region (South) and body mass index (higher BMI in the non-PK deficiency cohort)

Table 1. Patient characteristics

	PK deficiency cohort (N = 18)	Matched non-PK deficiency cohort (N = 90)
Age at index year, mean ± SD [median]	56.8 ± 13.6 [59.0]	56.8 ± 13.1 [59.0]
Category, years, n (%)		
> 20 to ≤ 30	1 (5.6)	5 (5.6)
> 30 to ≤ 40	0 (0.0)	0 (0.0)
> 40 to ≤ 50	5 (27.8)	25 (27.8)
> 50 to ≤ 60	4 (22.2)	20 (22.2)
> 60 to ≤ 70	7 (38.9)	35 (38.9)
> 70 to ≤ 80	0 (0.0)	0 (0.0)
> 80	1 (5.6)	5 (5.6)
Male, n (%)	17 (94.4)	85 (94.4)
US region, n (%)		
South	6 (33.3)	6 (6.7)
Midwest	2 (11.1)	4 (4.4)
North East	7 (38.9)	48 (53.3)
West	3 (16.7)	32 (35.6)
White, n (%)	15 (83.3)	77 (85.6)
Weight (lbs), mean ± SD [median]	190.0 ± 42.4 [189.2]	206.2 ± 47.5 [195.0]
Height (inches), mean ± SD [median]	69.3 ± 2.8 [69.9]	69.0 ± 2.9 [69.9]
BMI, mean ± SD [median]	27.6 ± 5.0 [26.7]	30.5 ± 6.7 [28.8]
20–25, n (%)	7 (38.9)	11 (12.2)
26–30, n (%)	5 (27.8)	37 (41.1)
> 30, n (%)	6 (33.3)	39 (43.3)
Charlson comorbidity index score, mean ± SD [median]	0.4 ± 1.2 [0]	0.5 ± 1.2 [0]

BMI = body mass index; PK = pyruvate kinase; SD = standard deviation.

Survival analysis

- The number of observed deaths over the follow-up period, and years until death for both cohorts are highlighted in **Table 2**

Table 2. Differences in observed deaths and years until death between cohorts

	PK deficiency cohort (N = 18)	Non-PK deficiency cohort (N = 90)
Years of follow-up, mean ± SD [median]	7.3 ± 5.2 [6.0]	9.2 ± 5.8 [8.0]
Observed deaths over follow-up period, n (%)	9 (50%)	28 (31%)
Years until death, median	10.9	17.1

PK = pyruvate kinase; SD = standard deviation.

RESULTS (CONTINUED)

- Patients in the non-PK deficiency cohort had a significantly longer time to death than the PK deficiency cohort (hazard ratio: 2.3; p = 0.0306; **Figure**)
- 10 years after index, 42% of patients in the PK deficiency cohort had died compared with 28% of those in the non-PK deficiency cohort

Figure. Kaplan–Meier survival estimates

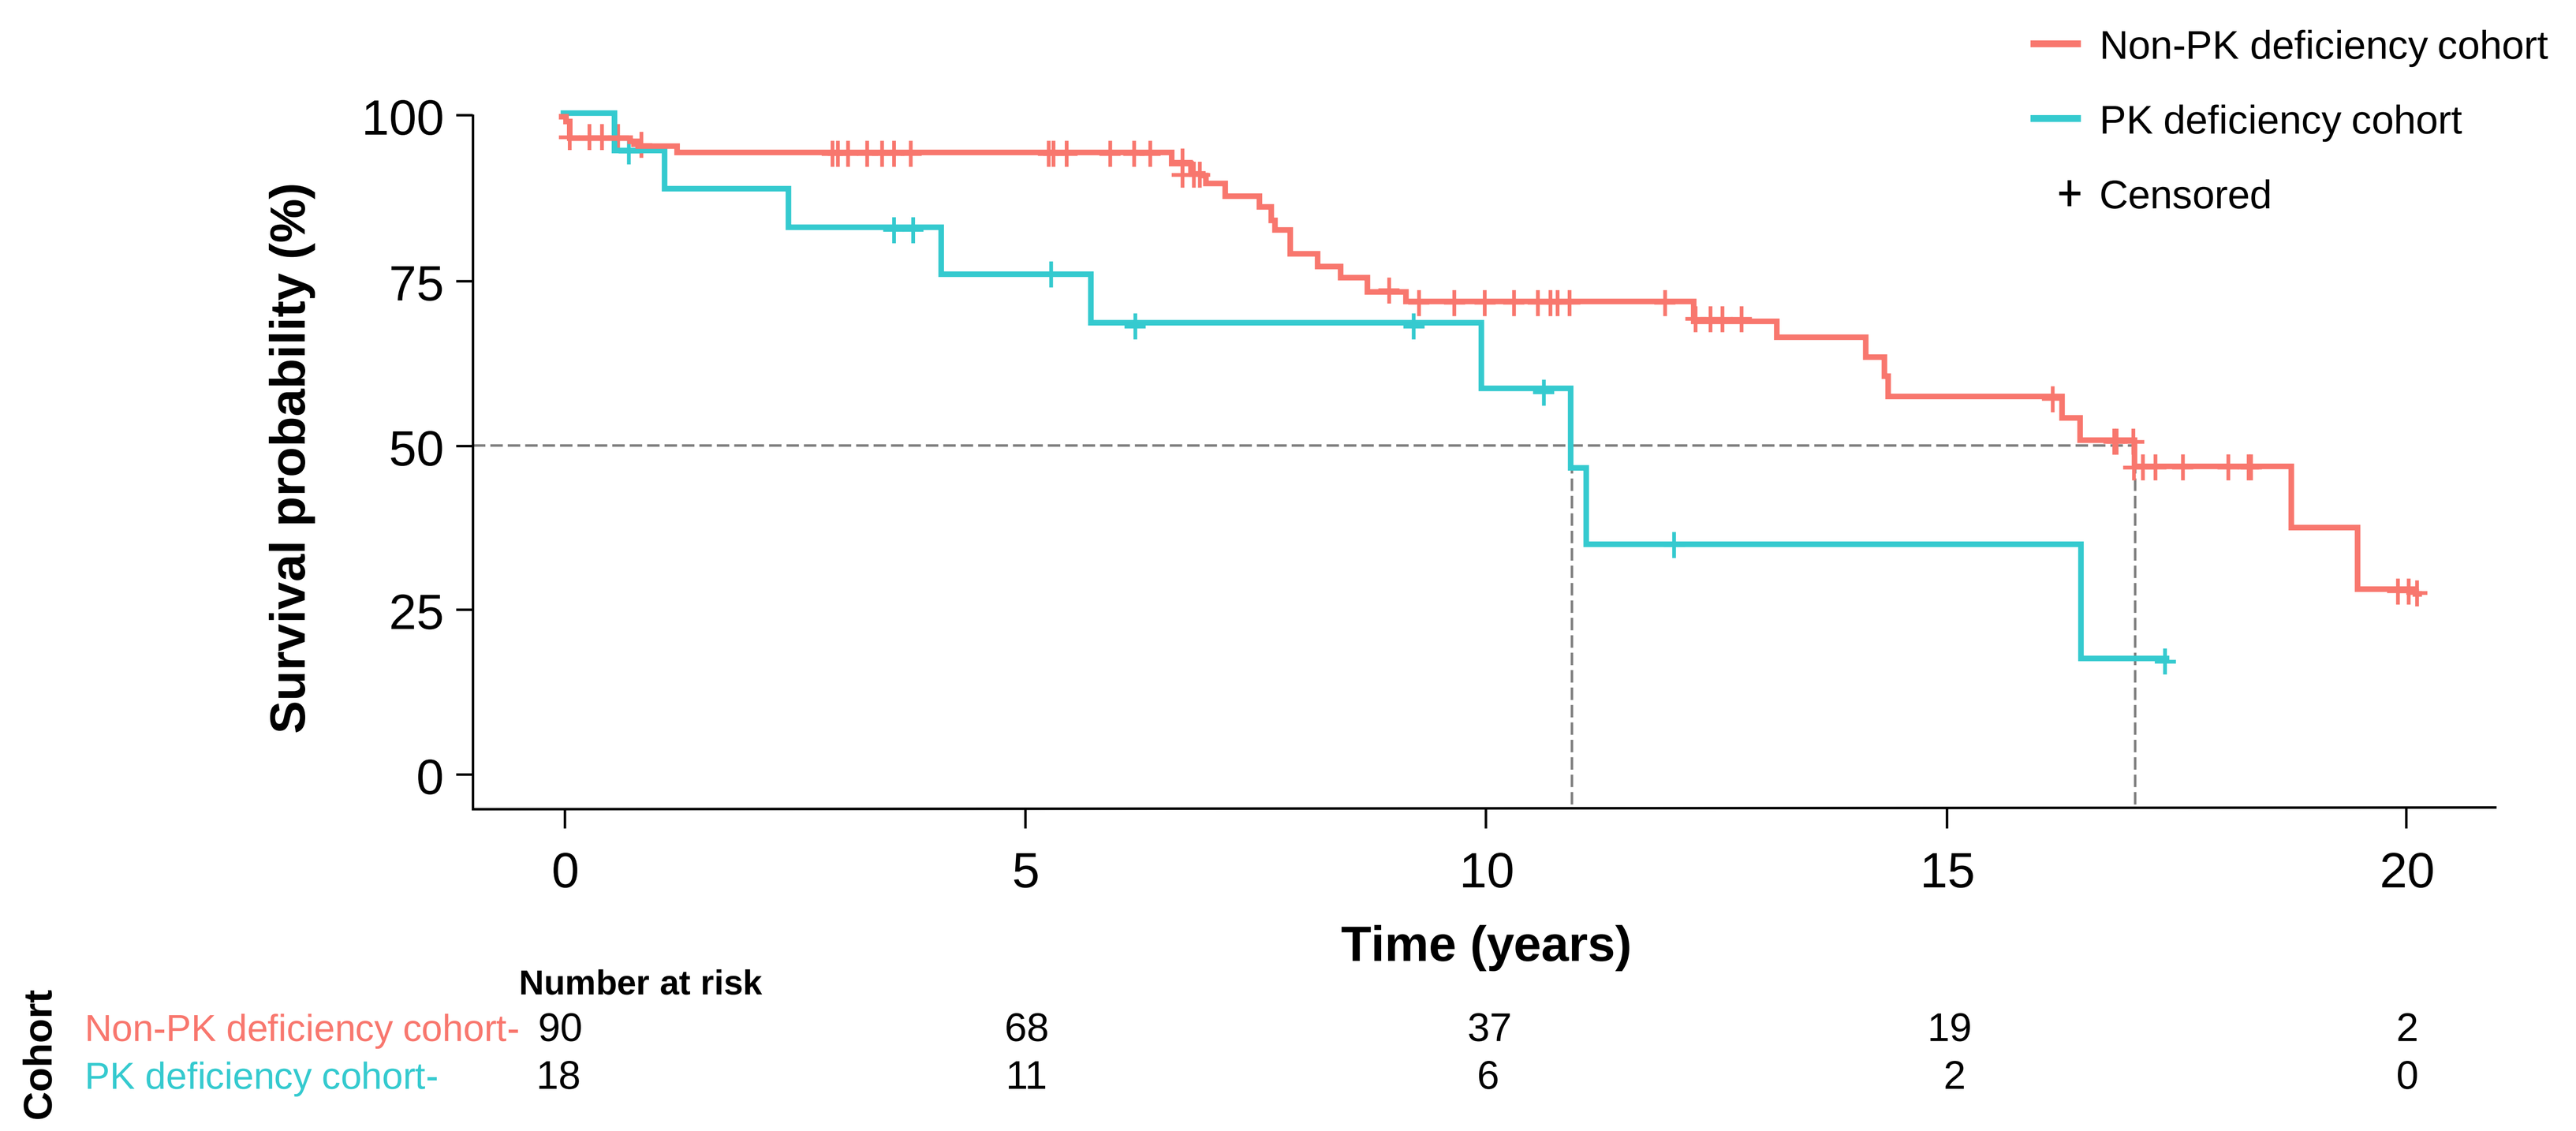
Non-PK deficiency cohort
PK deficiency cohort
+ Censored
Survival probability (%)
100
75
50
25
0
0
5
10
15
20
Time (years)
Number at risk
Cohort
Non-PK deficiency cohort- 90 68 37 19 2
PK deficiency cohort- 18 11 6 2 0

PK = pyruvate kinase.

Study strengths:

- The patients in the PK deficiency cohort had a manually-confirmed, physician-documented diagnosis
- This study is the first to compare mortality between patients with (physician-documented) PK deficiency and the general population

Study limitations:

- Females, pediatric, and adolescent population are underrepresented in this study
- Due to the rare and heterogeneous nature of PK deficiency, results of this study may not be generalizable

CONCLUSIONS

- **The results of this study suggest that patients with PK deficiency may be at an increased risk of mortality**
- **Further research is warranted to:**
 - **Understand cause of death among patients with PK deficiency**
 - **Examine mortality using larger sample sizes and other real-world data sources that better represent females and younger age groups**

Acknowledgements
Editorial assistance was provided by Onyx Medica, London, UK, and supported by Agios Pharmaceuticals, Inc.

Disclosures
This study was funded by Agios Pharmaceuticals, Inc. Author conflict of interest disclosures as follows: **Erin Zagadailov** and **Audra N. Boscoe**: Agios – employees and shareholders; **Viviana Garcia-Horton**: employed by Analysis Group, Inc., which received payment from Agios for participation in this research; **Sherry Shi**: employed by Analysis Group, Inc., which received payment from Agios for participation in this research; **Shuqian Liu**: has a research contract and received consulting fees from Analysis Group, Inc., which received payment from Agios for participation in this research; **Lizheng Shi**: has a research contract and received consulting fees from Analysis Group, Inc., which received payment from Agios for participation in this research; **Dendy Macaulay**: employed by Analysis Group, Inc., which received payment from Agios for participation in this research.

References

1. Grace RF et al. *Am J Hematol* 2015;90:825–30. **2.** Zanella A et al. *Br J Haematol* 2005;130:11–25. **3.** Grace RF et al. *Blood* 2018;131:2183–92. **4.** van Beers EJ et al. *Haematologica* 2019;104:e51–e3. **5.** Boscoe AN et al. *Blood* 2019;134. **6.** Grace RF et al. *Eur J Haematol* 2018;101:758–65. **7.** Grace RF et al. *Br J Haematol* 2019;184:721–34. **8.** Alli N et al. *Hematology* 2008;13:369–72; **9.** Nagai H et al. *Intern Med* 1994;33:56–59. **10.** Pérez-Albert P et al. *An Pediatr (Barc)* 2018;88:106–7. **11.** Pissard S et al. *Br J Haematol* 2006;133:683–89. **12.** Raphaël MF et al. *Am J Hematol* 2007;82:1025–28. **13.** Rider NL et al. *Am J Hematol* 2011;86:827–34. **14.** Zahid MF et al. *Blood* 2017;130:2906. **15.** Zanella A et al. *BJH* 1993;83:485–90. **16.** Zanella A et al. *Blood Rev* 2007;21:217–31.

Presented at European Haematology Association (EHA) Virtual Congress; 9–17 June 2021